Efficacy and safety of subcutaneous trastuzumab and intravenous trastuzumab as part of adjuvant therapy for HER2-positive early breast cancer: Final analysis of the randomised, two-cohort PrefHer study

Xavier Pivot,[1] Sunil Verma,[2] Lesley Fallowfield,[3] Volkmar Müller,[4] Mikhail Lichinitser,[5] Valerie Jenkins,[3] Alfonso Sánchez Muñoz,[6] Zuzana Machackova,[7] Stuart Osborne,[8] Joseph Gligorov[9]

[1]University Hospital Jean Minjoz, INSERM 1098, Besançon, France; [2]Tom Baker Cancer Centre, Department of Oncology, University of Calgary, AB, Canada; [3]Sussex Health Outcomes Research & Education in Cancer (SHORE-C), Brighton and Sussex Medical School, University of Sussex, Falmer, UK; [4]Department of Gynecology, University Medical Center Hamburg-Eppendorf, Hamburg, Germany; [5]Department of Chemotherapy and Combined Therapy, N.N. Blokhin Cancer Research Center, Moscow, Russia; [6]Servicio de Oncología Médica, Hospital Universitario Virgen de la Victoria, Málaga, Spain; [7]Global Product Development/Medical Affairs Oncology (PDMAO), F. Hoffmann-La Roche Ltd, Basel, Switzerland; [8]PDMA Operations (Biometrics), F. Hoffmann-La Roche Ltd, Basel, Switzerland; [9]Medical Oncology Department, APHP-Tenon, IUC-UPMC, Sorbonne University, Paris, France

209P

Background

- A 600 mg fixed-dose manual injection of subcutaneous trastuzumab (Herceptin® SC [H SC], F. Hoffmann-La Roche Ltd, Basel, Switzerland), given via hand-held syringe from an H SC Vial, was approved following demonstrated non-inferiority compared with intravenous trastuzumab (Herceptin® [H IV], F. Hoffmann-La Roche Ltd) based on pathological complete response and serum trough concentration in the HannaH study.[1]
- The international, open-label, randomised, crossover PrefHer study (NCT01401166) revealed overwhelming patient preferences for H SC, given by single-use injection device (SID) or hand-held syringe from an H SC Vial, over H IV as adjuvant therapy for HER2-positive early breast cancer.[2,3]
 - — Preferences were due to 'time saving' and 'less pain/discomfort/side effects,' and there was a high preference for H SC irrespective of whether or not patients received H IV prior to study enrolment.[2,3]
- We present here the efficacy results and final safety data from the PrefHer study.

Methods

- Patients with histologically confirmed primary invasive breast adenocarcinoma with no evidence of residual, locally recurrent or metastatic disease after completion of surgery and neoadjuvant or adjuvant chemotherapy (with or without H) were randomised to receive either four cycles of H SC every 3 weeks (q3w), then four cycles of H IV, or the reverse sequence (Figure 1).

Figure 1. Overall study design[2]

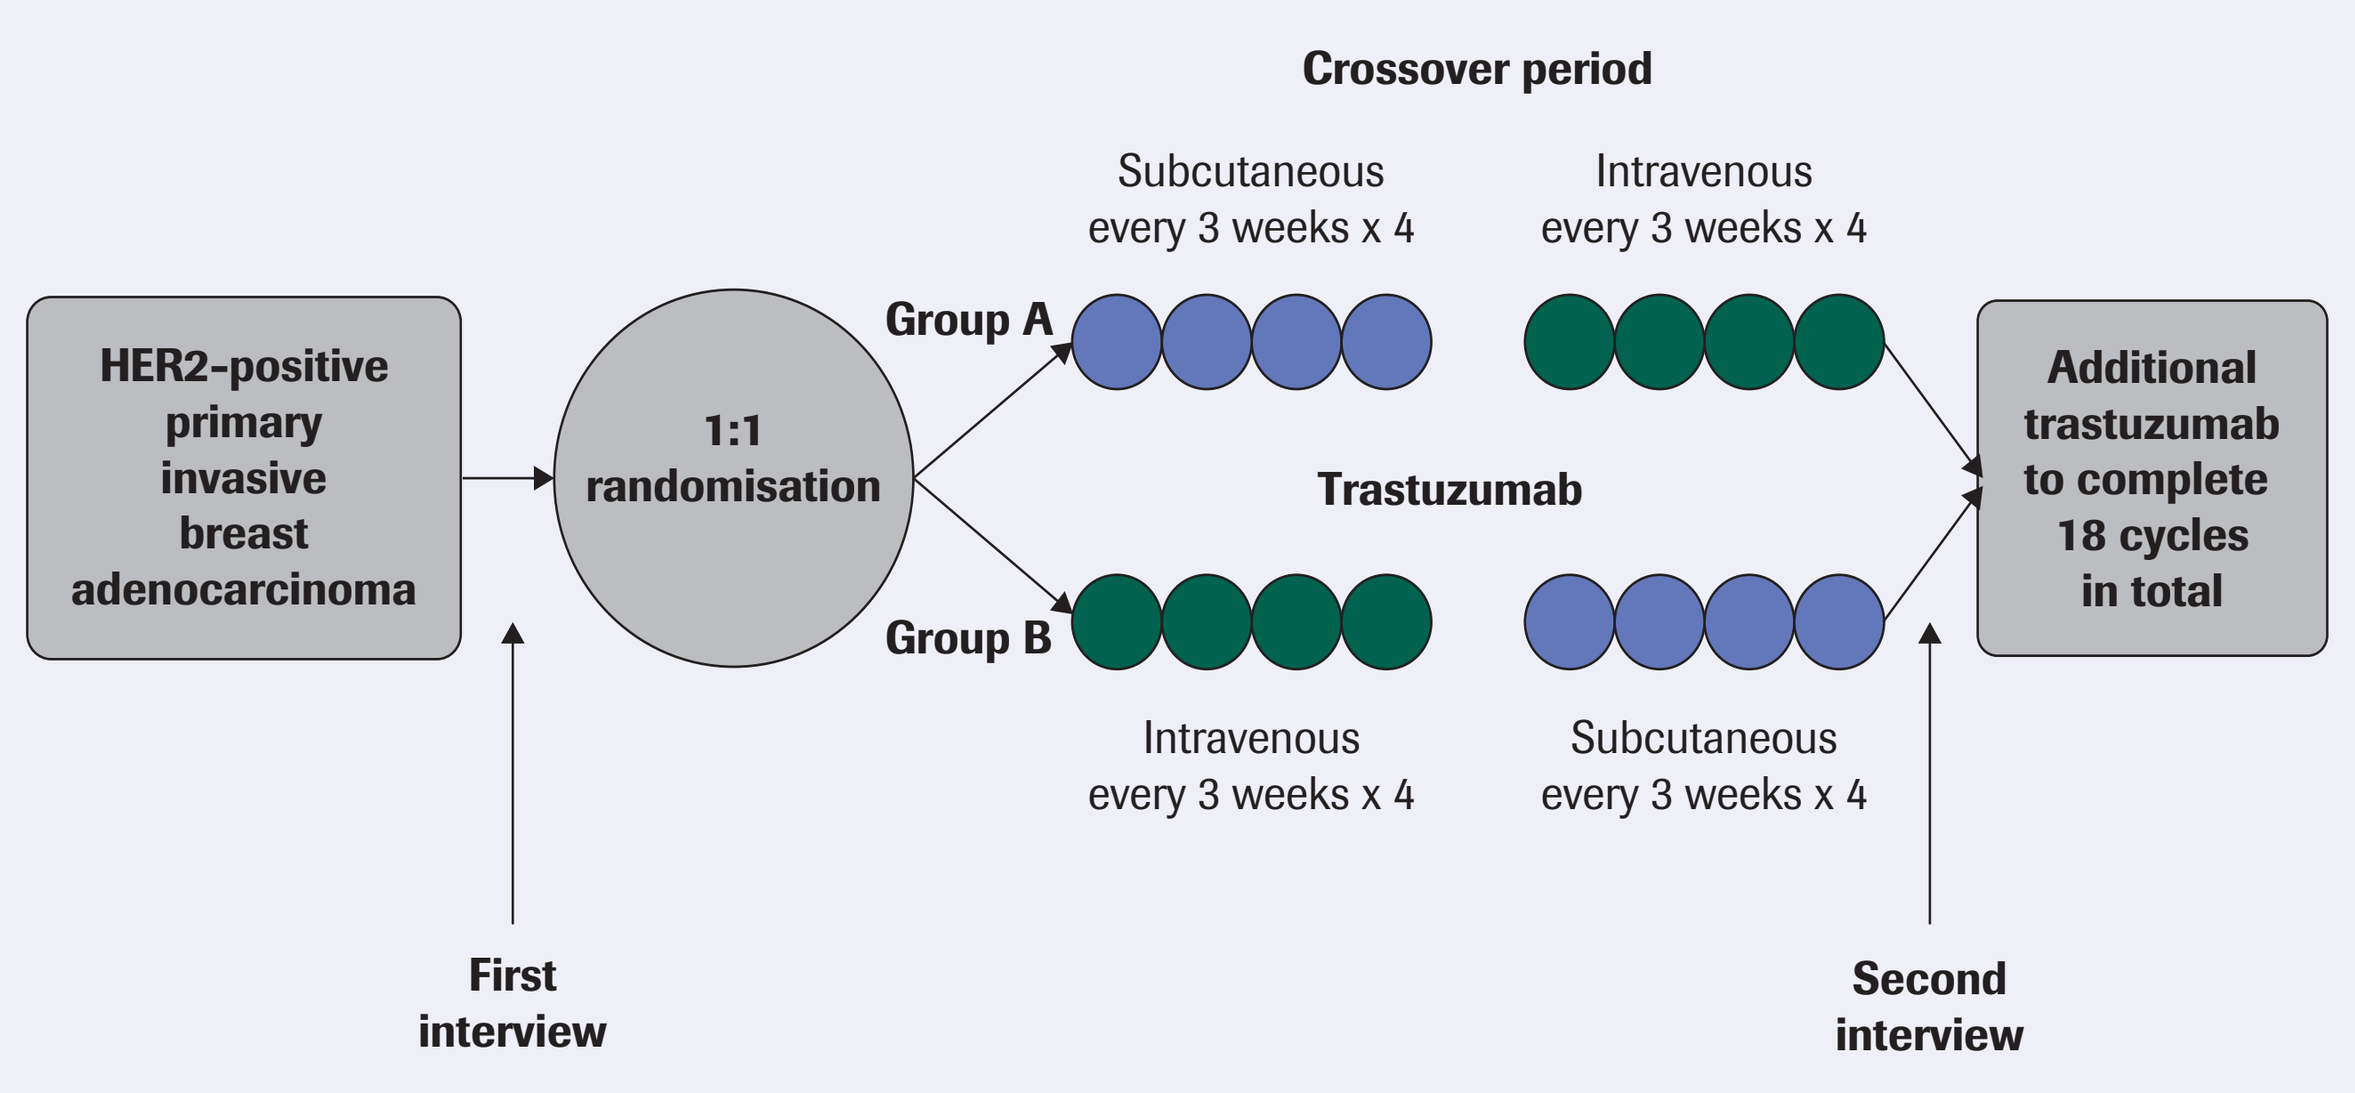

Crossover period
Subcutaneous every 3 weeks x 4
Intravenous every 3 weeks x 4
HER2-positive primary invasive breast adenocarcinoma
1:1 randomisation
Group A
Trastuzumab
Group B
Additional trastuzumab to complete 18 cycles in total
Intravenous every 3 weeks x 4
Subcutaneous every 3 weeks x 4
First interview
Second interview

Randomisation was stratified according to whether or not patients had already received intravenous trastuzumab.

Reprinted from *Lancet Oncology*, Vol 14, Pivot X, Gligorov J, Müller V, Barrett-Lee P, Verma S, Knoop A, Curigliano G, Semiglazov V, López-Vivanco G, Jenkins V, Scotto N, Osborne S, Fallowfield L; PrefHer Study Group, Preference for subcutaneous or intravenous administration of trastuzumab in patients with HER2-positive early breast cancer (PrefHer): an open-label randomised study, 962–970.

Copyright (2013), with permission from Elsevier. Licence Number: 3954200849209

- During the crossover period, patients in Cohort 1 received H SC via SID and patients in Cohort 2 received H SC via hand-held syringe from an H SC Vial. Following the crossover period, i.e., the H continuation period, it was planned for patients in Cohort 1 to receive H IV (unless participating in SID self-administration), and for patients in Cohort 2 to receive H SC via hand-held syringe from an SC Vial.
- Patients could have been either H-naive (*de novo*) or could have already started H treatment for early breast cancer prior to study entry (non-*de novo*), but needed to receive at least eight more cycles to complete 1 year (18 cycles) of H treatment in the adjuvant setting.
- The primary endpoint, patient preference for H SC or H IV, has been reported elsewhere as described in the Background section, along with safety data from the crossover period.[2,3] We report here a secondary endpoint, 3-year event-free survival (EFS), defined as time from randomisation to local, regional or distant disease recurrence, contralateral breast cancer or death from any cause, as well as safety across both groups of the study in the crossover and H continuation periods.
- EFS was assessed using the Kaplan–Meier approach and is shown for the overall evaluable intention-to-treat (ITT) populations of each cohort, and overall.
- Adverse events (AEs) and serious AEs (SAEs) were reported according to NCI-CTCAE v4 and New York Heart Association criteria.

Results

- Of 488 patients randomised, 483 received at least one dose of study treatment and were evaluated for safety.[3] A total of 467 patients who received at least one dose of H IV and H SC and completed patient interviews before randomisation and at the end of crossover treatment for the primary endpoint (patient preference) were included in the evaluable ITT population.[3]
- The *de novo* group comprised 98/483 patients (20.3%) and the non-*de novo* group 385/483 patients (79.7%).
- A total of 409 patients completed follow-up according to protocol. Baseline characteristics were balanced between treatment groups, as previously described.[3]

Treatment exposure

- Taking into account the H cycles received prior to randomisation, a total of 425/483 (88.0%) patients in the safety population received all 18 cycles of H, with a median of 13 cycles on-study.
- The majority of patients in the *de novo* group (89/98, 90.8%) completed all 18 cycles of H.
- Taking into account H cycles received before randomisation, the majority of non-*de novo* patients (336/385, 87.3%) also completed all 18 cycles of H.

Event-free survival

- After median follow-up of 36.1 months, 3-year EFS across both groups in the evaluable ITT population was 90.6% overall (Figure 2A), 89.9% in Cohort 1 (Figure 2B) and 91.1% in Cohort 2 (Figure 2C).

Figure 2. EFS in the evaluable ITT population (both study groups combined)

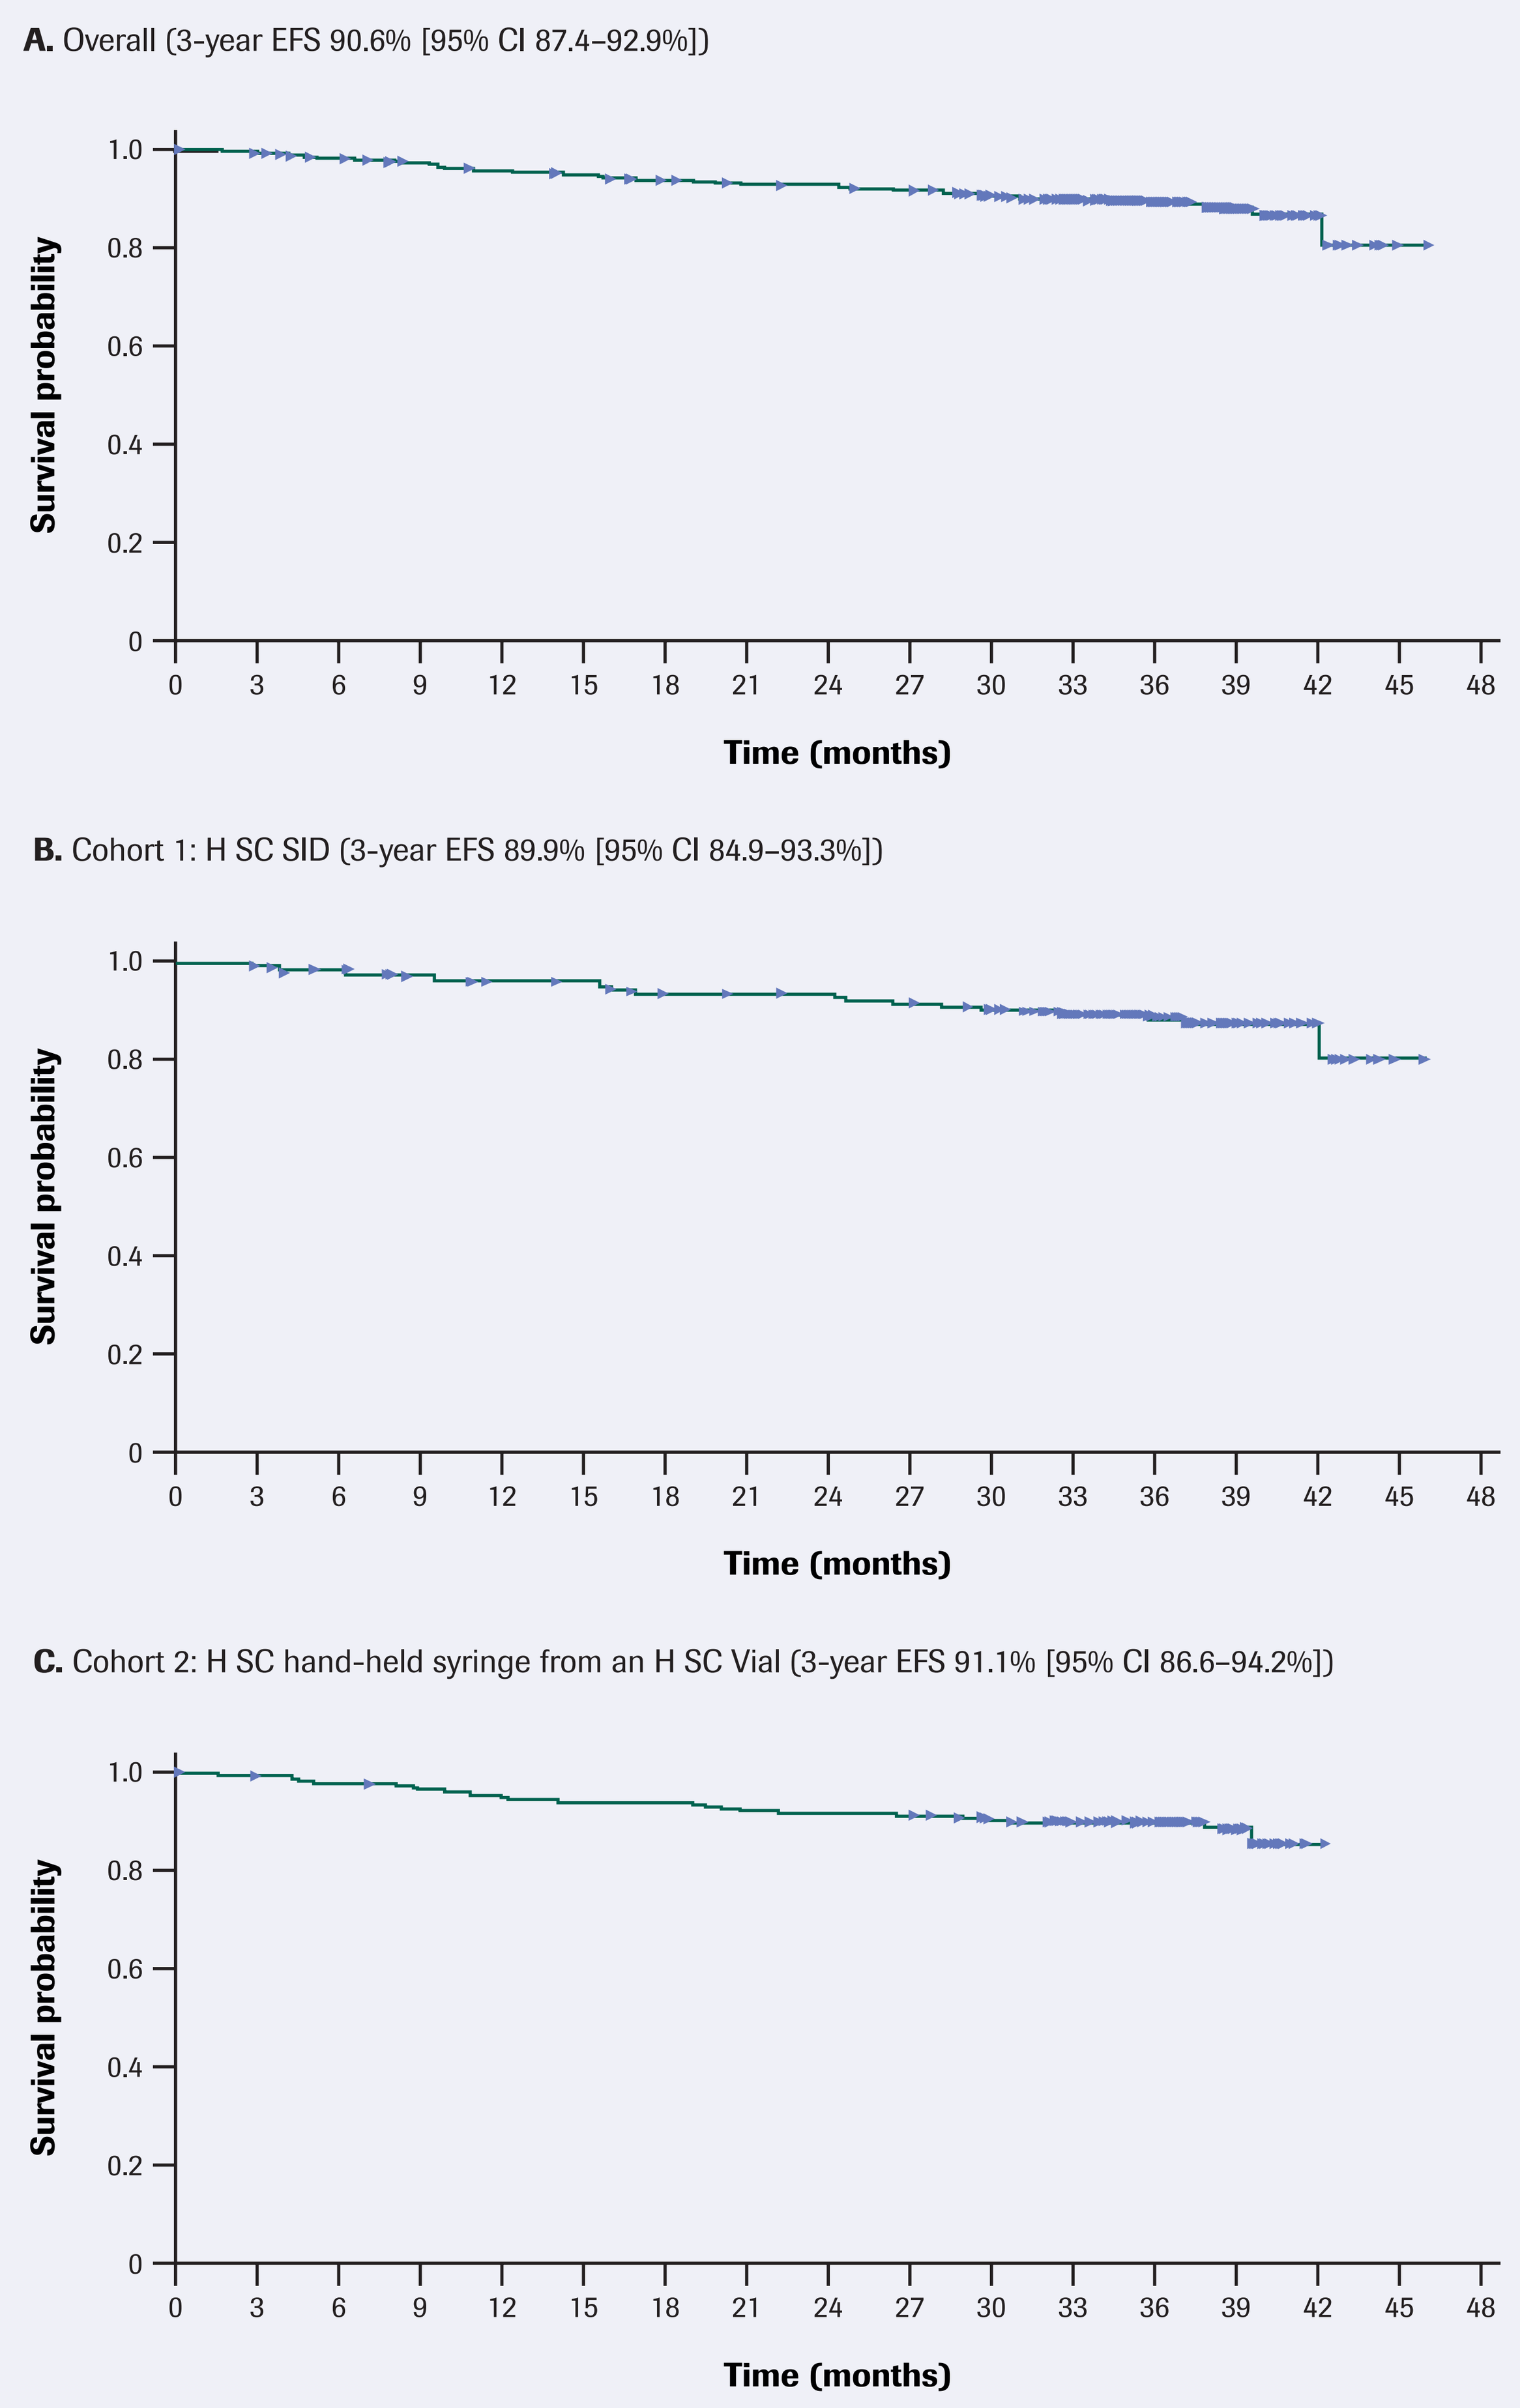

A. Overall (3-year EFS 90.6% [95% CI 87.4–92.9%])
B. Cohort 1: H SC SID (3-year EFS 89.9% [95% CI 84.9–93.3%])
C. Cohort 2: H SC hand-held syringe from an H SC Vial (3-year EFS 91.1% [95% CI 86.6–94.2%])
Survival probability
1.0
0.8
0.6
0.4
0.2
0
0 3 6 9 12 15 18 21 24 27 30 33 36 39 42 45 48
Time (months)

EFS is defined as time from randomisation to local, regional or distant disease recurrence, contralateral breast cancer or death from any cause.

CI, confidence interval; EFS, event-free survival; H, trastuzumab (Herceptin®); SC, subcutaneous; SID, single-use injection device.

Safety

- Safety by treatment period is shown in Table 1.

Table 1. Summary of safety by treatment period (both cohorts combined)

Patients with ≥ 1 AE, n (%)*	H SC period during crossover n = 479	H IV period during crossover n = 478	H continuation n = 440	H SC SID self-administration during H continuation n = 43	Overall n = 483
Median H cycles, n	4.0	4.0	5.0	2.0	13.0
Any AE	300 (62.6)	258 (54.0)	223 (50.7)	12 (27.9)	388 (80.3)
Grade 1	262 (54.7)	206 (43.1)	175 (39.8)	10 (23.3)	360 (74.5)
Grade 2	119 (24.8)	110 (23.0)	85 (19.3)	5 (11.6)	214 (44.3)
Grade 3	17 (3.5)	16 (3.3)	16 (3.6)	1 (2.3)	45 (9.3)
Grade 4	0	0	0	0	0
Grade 5	0	0	0	0	0
Treatment-related AE	163 (34.0)	53 (11.1)	60 (13.6)	4 (9.3)	207 (42.9)
Discontinuation for AE	5 (1.0)	6 (1.3)	10 (2.3)	0	21 (4.3)
Any SAE	4 (0.8)	4 (0.8)	11 (2.5)	1 (2.3)	19 (3.9)
Treatment-related SAE	0	0	1 (0.2)	0	1 (0.2)

* Could be counted once per grade but ≥ once overall.

AE, adverse event; H, trastuzumab (Herceptin®); IV, intravenous; SAE, serious adverse event; SC, subcutaneous; SID, single-use injection device.

- The most common AEs across both cohorts (all grades) were arthralgia (13.7%), asthenia (13.7%) and headache (10.4%), with no other AEs occurring in ≥ 10% of patients.
 - — Differences in AE rates between H SC and H IV periods during crossover were driven by injection site reactions, and rates were comparable between H SC and H IV periods when injection site reactions were excluded.
- Most AEs were Grade 1 or 2, with Grade 3 AEs in 45 patients (9.3%). No Grade 3 AE occurred in more than 1% of patients. There were no Grade 4 or 5 AEs.
- AEs considered by the investigator to be related to H treatment were reported in 213 patients (44.1%), and at Grade 3 severity in 14 patients (2.9%). Left ventricular dysfunction and dyspnoea (two patients each) were the only H-related Grade 3 AEs that occurred in more than one patient.
- SAEs were reported in 19 patients (3.9%). Only one of 19 (left ventricular dysfunction in one patient) was considered by the investigator to be related to H treatment. All SAEs had resolved by clinical cut-off.

Cardiac events

- Cardiac AEs are summarised in Table 2.
- Most were Grade 1 or 2, and only one serious cardiac AE was reported (left ventricular dysfunction, above).
- Only four patients had Grade 3 cardiac AEs (three left ventricular dysfunction, one congestive heart failure).
- Congestive heart failure was reported in five patients (1.0%), none serious.

Table 2. Cardiac AEs

Patients with ≥ 1 AE, n (%)*	H SC period during crossover n = 479	H IV period during crossover n = 478	H continuation n = 440	H SC SID self-administration during H continuation n = 43	Overall n = 483
Any cardiac AE	12 (2.5)	15 (3.1)	17 (3.9)	0	40 (8.3)
Grade 1	9 (1.9)	11 (2.3)	11 (2.5)	0	28 (5.8)
Grade 2	2 (0.4)	3 (0.6)	6 (1.4)	0	10 (2.1)
Grade 3	1 (0.2)	2 (0.4)	1 (0.2)	0	4 (0.3)
Cardiac disorders†	8 (1.7)	14 (2.9)	14 (3.2)	0	33 (6.8)
Left ventricular dysfunction	2 (0.4)	5 (1.0)	4 (0.9)	0	11 (2.3)
Palpitations	3 (0.6)	2 (0.4)	2 (0.5)	0	7 (1.4)
Congestive heart failure	2 (0.4)	0	3 (0.7)	0	5 (1.0)
Investigations‡	4 (0.8)	3 (0.6)	3 (0.7)	0	9 (1.9)

* Could be counted once per grade but ≥ once overall.

† Cardiac disorders not listed: bradycardia (three patients), extrasystoles (two patients), angina pectoris, cardiomyopathy, diastolic dysfunction, heart valve incompetence, left ventricular hypertrophy, mitral valve incompetence, sinus bradycardia, tachycardia (one patient each).

‡ Ejection fraction decreased (seven patients), ejection fraction abnormal, electrocardiogram change (one patient each).

AE, adverse event; H, trastuzumab (Herceptin®); IV, intravenous; SC, subcutaneous; SID, single-use injection device.

Conclusions

- Overall, the 3-year EFS rates observed in both cohorts of the PrefHer study were consistent with those observed in previous clinical trials of H therapy for patients with HER2-positive early breast cancer.[4–8]
- No new safety signals were identified with longer follow-up, with only one cardiac SAE reported in 483 patients.
- The overall safety profile during adjuvant treatment was as expected.
- Consistent results from both cohorts of PrefHer, combined with data from the HannaH study, demonstrate that H SC is a valid and well tolerated option for patients and healthcare professionals, regardless of H SC delivery method (SID or hand-held syringe from an H SC Vial).

Acknowledgements

We thank the individuals who contributed to the design of the study instruments, the patients, their families, the nurses, the interviewers and the investigators who participated in this study. Funding for the PrefHer study was provided by F. Hoffmann-La Roche Ltd, Basel, Switzerland. Support for third-party writing assistance for this poster, furnished by John Carron PhD, of Health Interactions, was provided by F. Hoffmann-La Roche Ltd, Basel, Switzerland.

References

1. Ismael G, *et al. Lancet Oncol* 2012; **13**:869–878; 2. Pivot X, *et al. Lancet Oncol* 2013; **14**:962–970; 3. Pivot X, *et al. Ann Oncol* 2014; **25**:1979–1987; 4. Romond EH, *et al. N Engl J Med* 2005; **353**:1673–1684; 5. Perez EA, *et al. J Clin Oncol* 2011; **29**:3366–3373; 6. Slamon D, *et al. N Engl J Med* 2011; **365**:1273–1283; 7. Pivot X, *et al. Lancet Oncol* 2013; **14**:741–748; 8. Jackisch C, *et al. Eur J Cancer* 2016; **62**:62–75.

PUSHED FOR TIME?

In order to use the QR code, please use or download an app called 'QR code reader' from the Apple Appstore or the Android Playstore

Receive an instant copy of this poster

Request additional presentations of trials sponsored/supported by Roche at this congress

NB: There may be associated costs for downloading data. These costs may vary depending on your service provider and may be high if you are using your smartphone abroad. Please check your phone tariff or contact your service provider for more details. Copies of this poster obtained through QR (Quick Response) code are for personal use only and may not be reproduced without written permission of the authors.

Contact the lead author at xavier.pivot@univ-fcomte.fr for permission to reprint and/or distribute.

Presented at the European Society for Medical Oncology 2016 Congress, 7–11 October, Copenhagen, Denmark

2025723 ESMO 2016 PrefHer Final Analyses Poster v8.indd 1

04/10/2016 14:22